P02-05

Mitigating effect of EC-18 on influenza A (H1N1) virus-induced acute respiratory distress syndrome and SARS-CoV-2 infection *in vitro* and *in vivo*

Yong-Jae Kim[1], Jinseon Jeong[1], Ki-Young Sohn[1], Kaapjoo Park[1], Sun Woo Yoon[2], Sun Young Yoon[1]* and Jae Wha Kim[2]*

1Enzychem Lifesciences Corporation, 2Biomedical Translational Research Center, KRIBB, Daejeon 305-806, Republic of Korea.

ABSTRACT

Since the 2009 H1N1 pandemic, Influenza A (H1N1) virus has become a seasonal virus circulating worldwide. Additionally, severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2), which causes 2019 COVID-19 pandemic, has rapidly spread worldwide. These viruses are one of the most important causes of acute lung injury (ALI) and acute respiratory distress syndrome (ARDS), which result in high mortality in elderly patients with underlying degenerative diseases. EC-18 (1-palmityoyl-2-linoleoyl-3-acetyl-rac-glycerol) is a synthetic mono-acetyldiglyceride(MW=635) currently under development as an oral agent to prevent or to mitigate the virus-induced ALI and ARDS. In this study, we established a murine model of viral infection by infecting C57BL/6 mice with a sub-lethal dose of H1N1 [PR8] influenza A virus (10^3 $TCID_{50}$) to evaluate the efficacy of EC-18 on histopathology, immune cell infiltration, cytokine levels and virus titers in the pulmonary region at various times post-inoculation. Mice received 50 mg/kg of EC-18 via oral gavage 1 h prior to virus inoculation and continued daily administration until the end of experiment. H1N1 virus has significantly caused body-weight loss, histological impairment in the lungs and elevated immune cell infiltration and cytokine levels in broncho-alveolar lavage fluid (BALF). EC-18 effectively ameliorated the body-weight loss and virus titers in lungs of the infected mice. Also, EC-18 attenuated H1N1-induced histological changes in the lungs, and reduced immune cell infiltration and the cytokine levels in BALF. Moreover, EC-18 effectively decreased extracellular HMGB1 released from dying bronchial and alveolar cells. Similar to the EC-18 effect on H1N1 virus, we investigated the effects of EC-18 on SARS-CoV-2-induced cytopathic effect (CPE) and viral replication in Vero E6 cells. As a result, EC-18 significantly and dose-dependently inhibited SARS-CoV-2-mediated CPE and viral replication (about 82.5 and 99.8% inhibition of viral replication). Based on these observations, we strongly believe that EC-18 has high potential as an anti-viral agent against H1N1 and SARS-CoV-2 infections.

INTRODUCTION

- Influenza A (H1N1) or SARS-CoV-2 virus infection causes a highly contagious disease that leads to inflammatory lung injury and pneumonia. The virus infection-induced pulmonary damage and inflammation is correlated with morbidity and mortality. **[Andre C. Kalil. et al. Critical Care volume 23, Article number: 258 (2019)] [Weiqi Hong. et al. Signal Transduction and Targeted Therapy (2021) 6:1]**
- EC-18 is a synthetic mono-acetyl-diacylglyceride, and an effective immune regulator in diverse inflammatory disease model. **[Yoon, S. Y., et al. (2015). Immune Netw 15(2): 100-109.]**

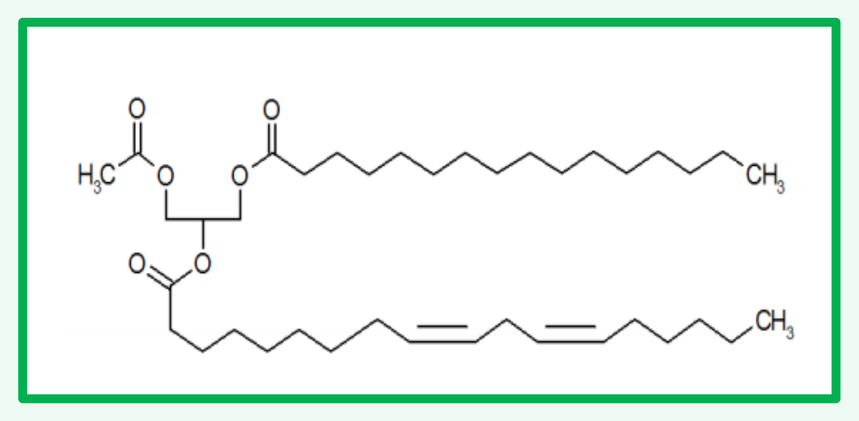

EC-18

- 1-Palmitoyl-2-linoleoyl-3-acetyl-rac-glycerol
- C39H70O6
- MW: 657.9

Experimental Design

1. A mouse model of H1N1 Influenza A virus infection

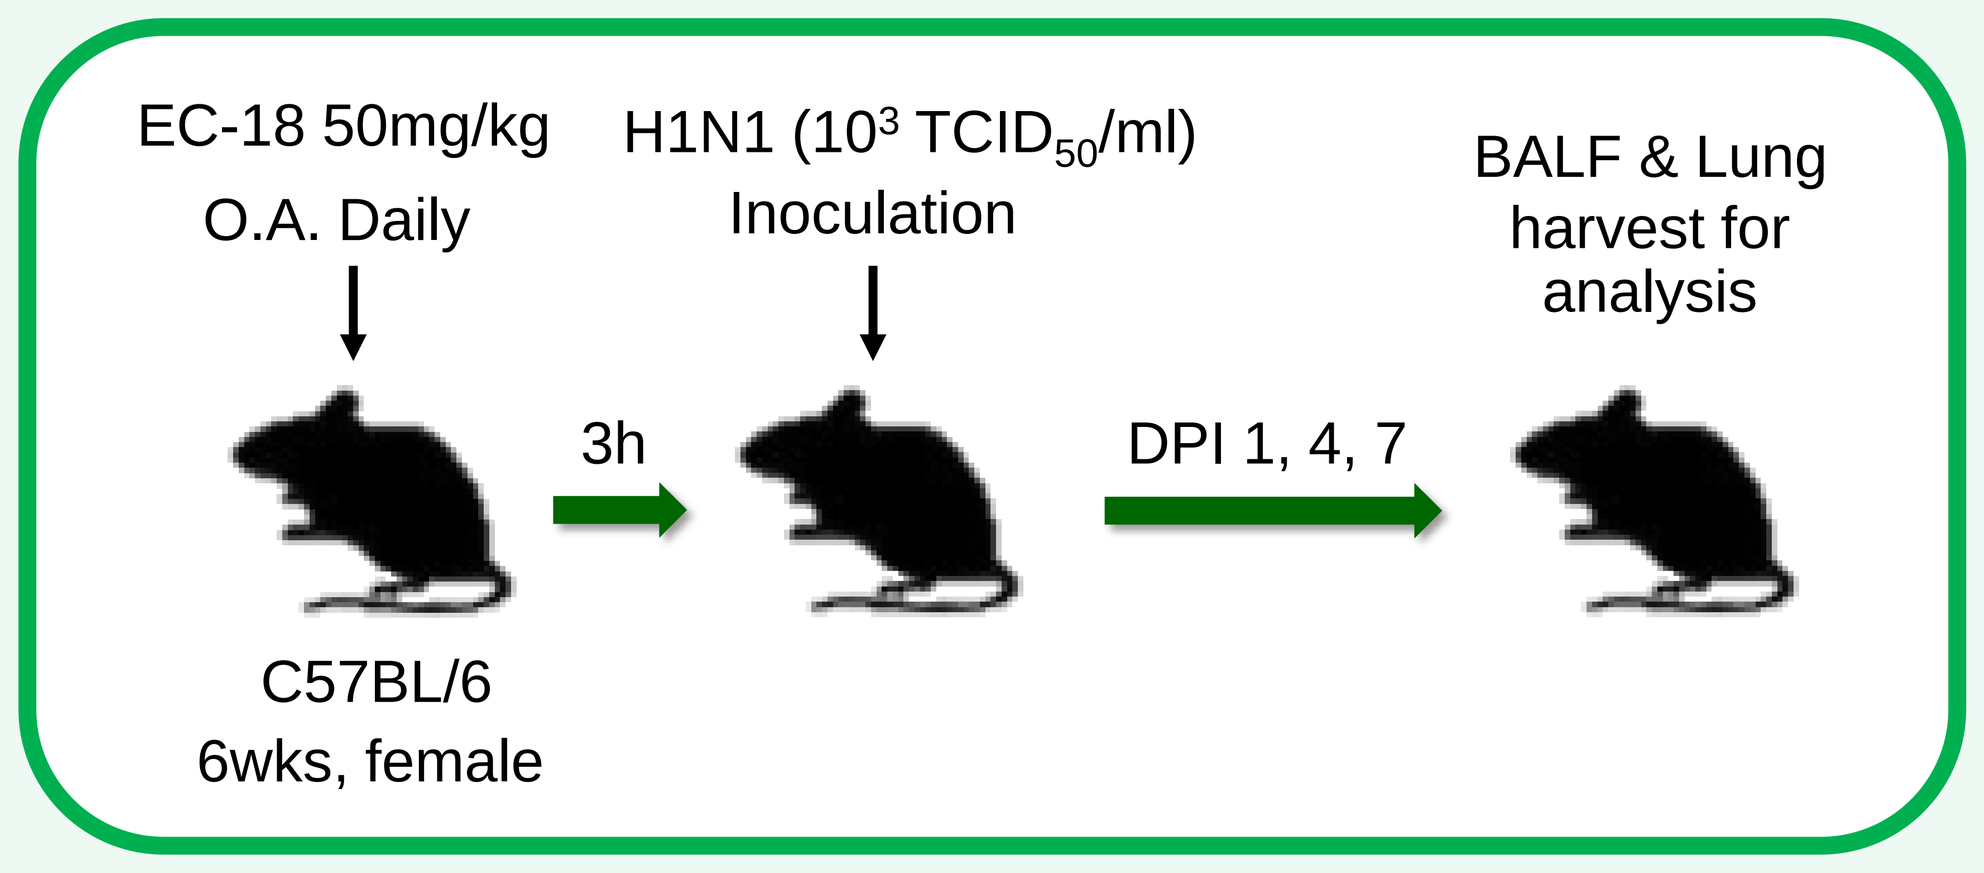
EC-18 50mg/kg
O.A. Daily
H1N1 (10^3 $TCID_{50}$/ml)
Inoculation
BALF & Lung harvest for analysis
3h
DPI 1, 4, 7
C57BL/6
6wks, female

2. A mouse model of SARS-CoV-2 virus infection

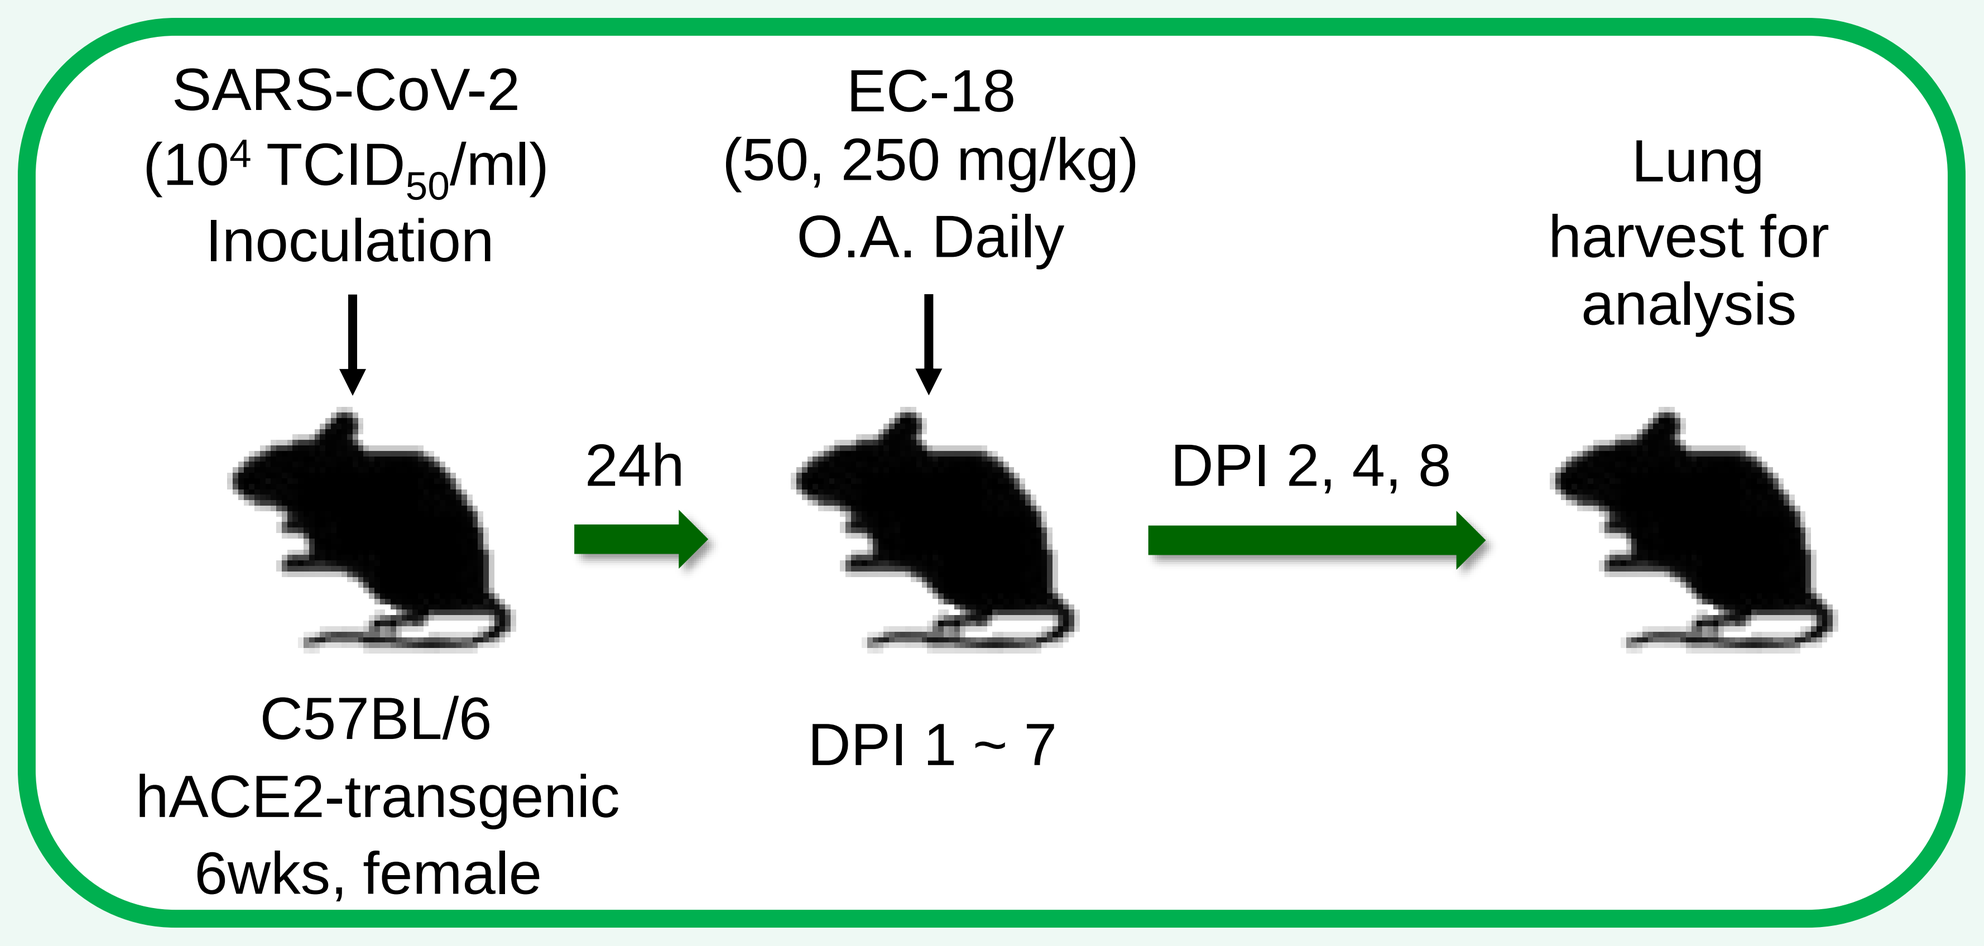
SARS-CoV-2
(10^4 $TCID_{50}$/ml)
Inoculation
EC-18
(50, 250 mg/kg)
O.A. Daily
Lung harvest for analysis
24h
DPI 2, 4, 8
C57BL/6
hACE2-transgenic
6wks, female
DPI 1 ~ 7

Conclusion

- EC-18 mitigated H1N1 IAV-induced weight loss and inflammatory lung injury by decreasing chemokine expression, HMGB1 release and inflammatory cell infiltration.
- Also, EC-18 inhibits virus replication in lung of infected mice.
- EC-18 mitigated SARS-CoV-2 virus-induced pneumonia by down-regulating virus replication.
- Our results show therapeutic potential of EC-18 for preventing virus-induced inflammatory lung injury.

RESULT 1 : H1N1 Influenza A Virus

1. EC-18 mitigates body weight reduction in H1N1 influenza A virus (IAV)-infected mice.

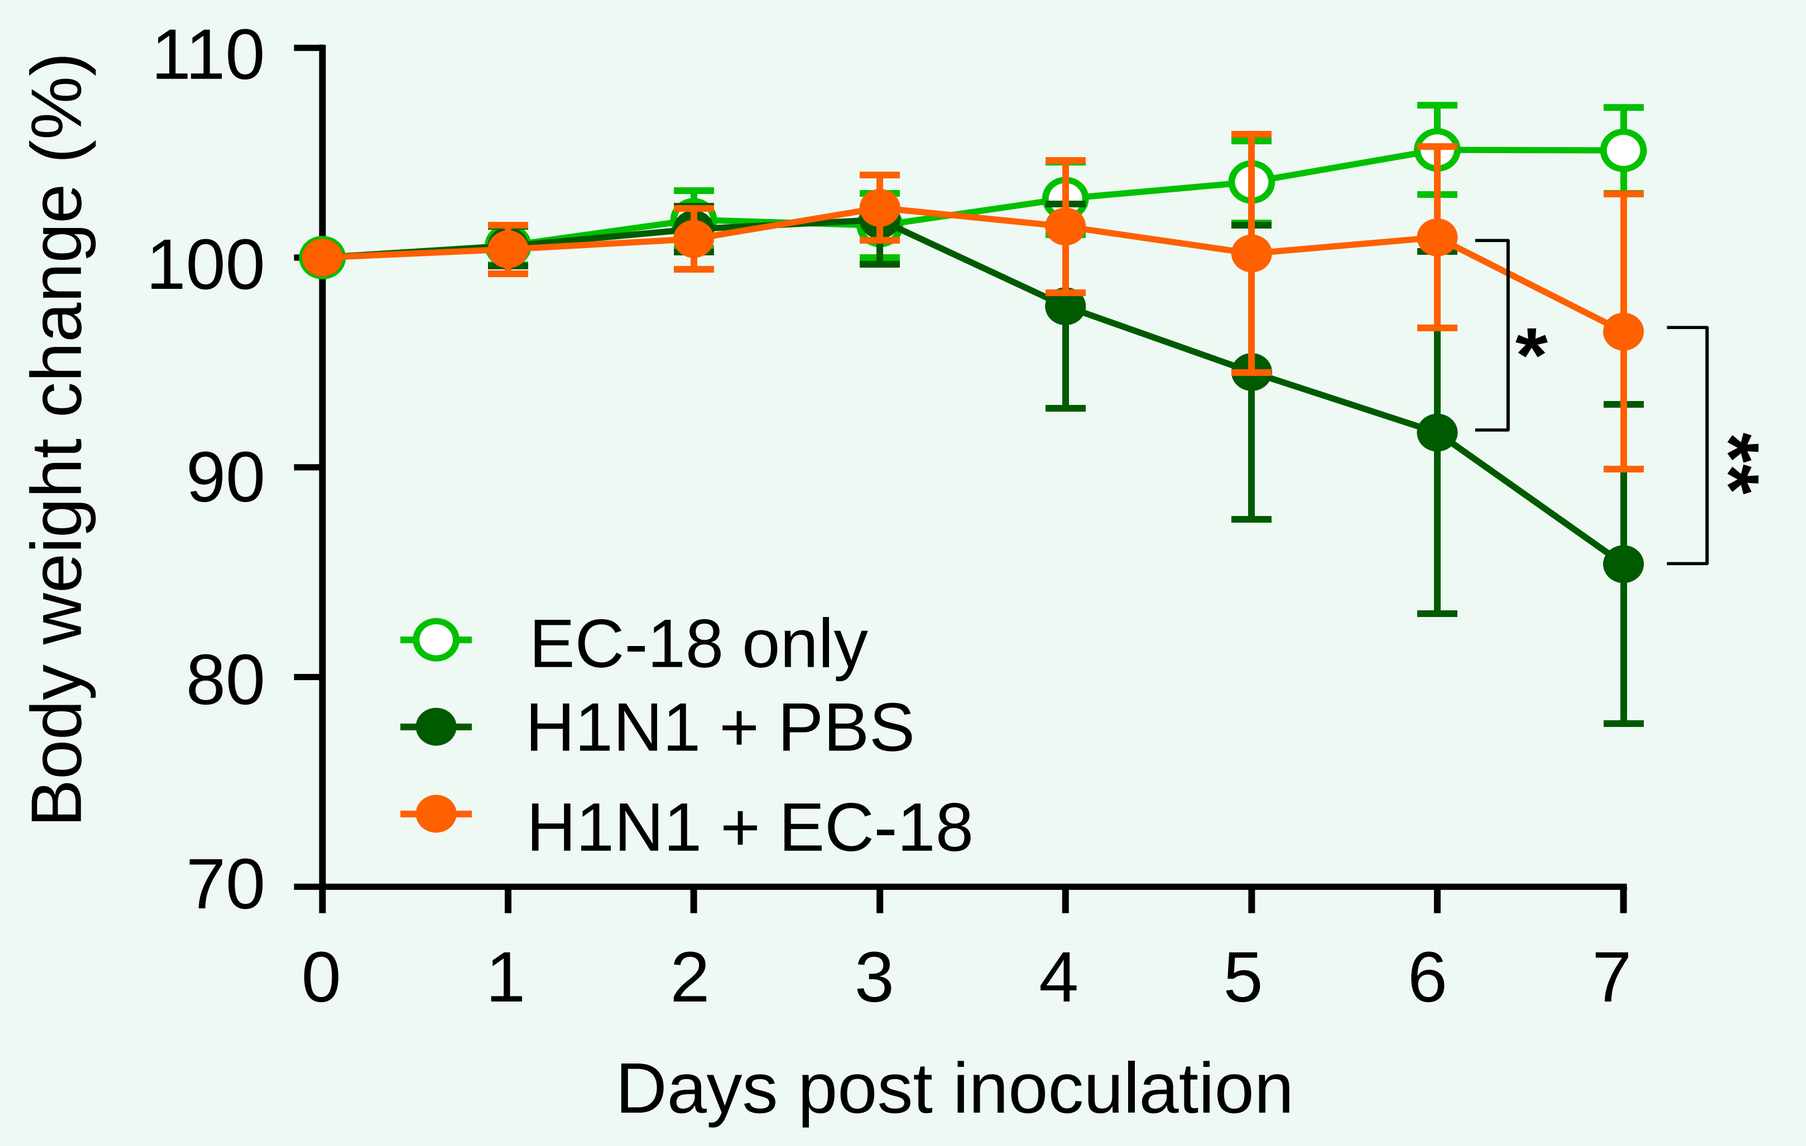
Body weight change (%)
EC-18 only
H1N1 + PBS
H1N1 + EC-18
Days post inoculation

Body weight change of mice infected with the H1N1 influenza A virus. The mice were infected with a sub-lethal dose of H1N1 IAV, and body weights were monitored everyday. (n=8-9) $*p < 0.05$, $**p < 0.01$ vs. H1N1 + PBS.

2. EC-18 attenuates virus titer in lungs of IAV-infected mice.

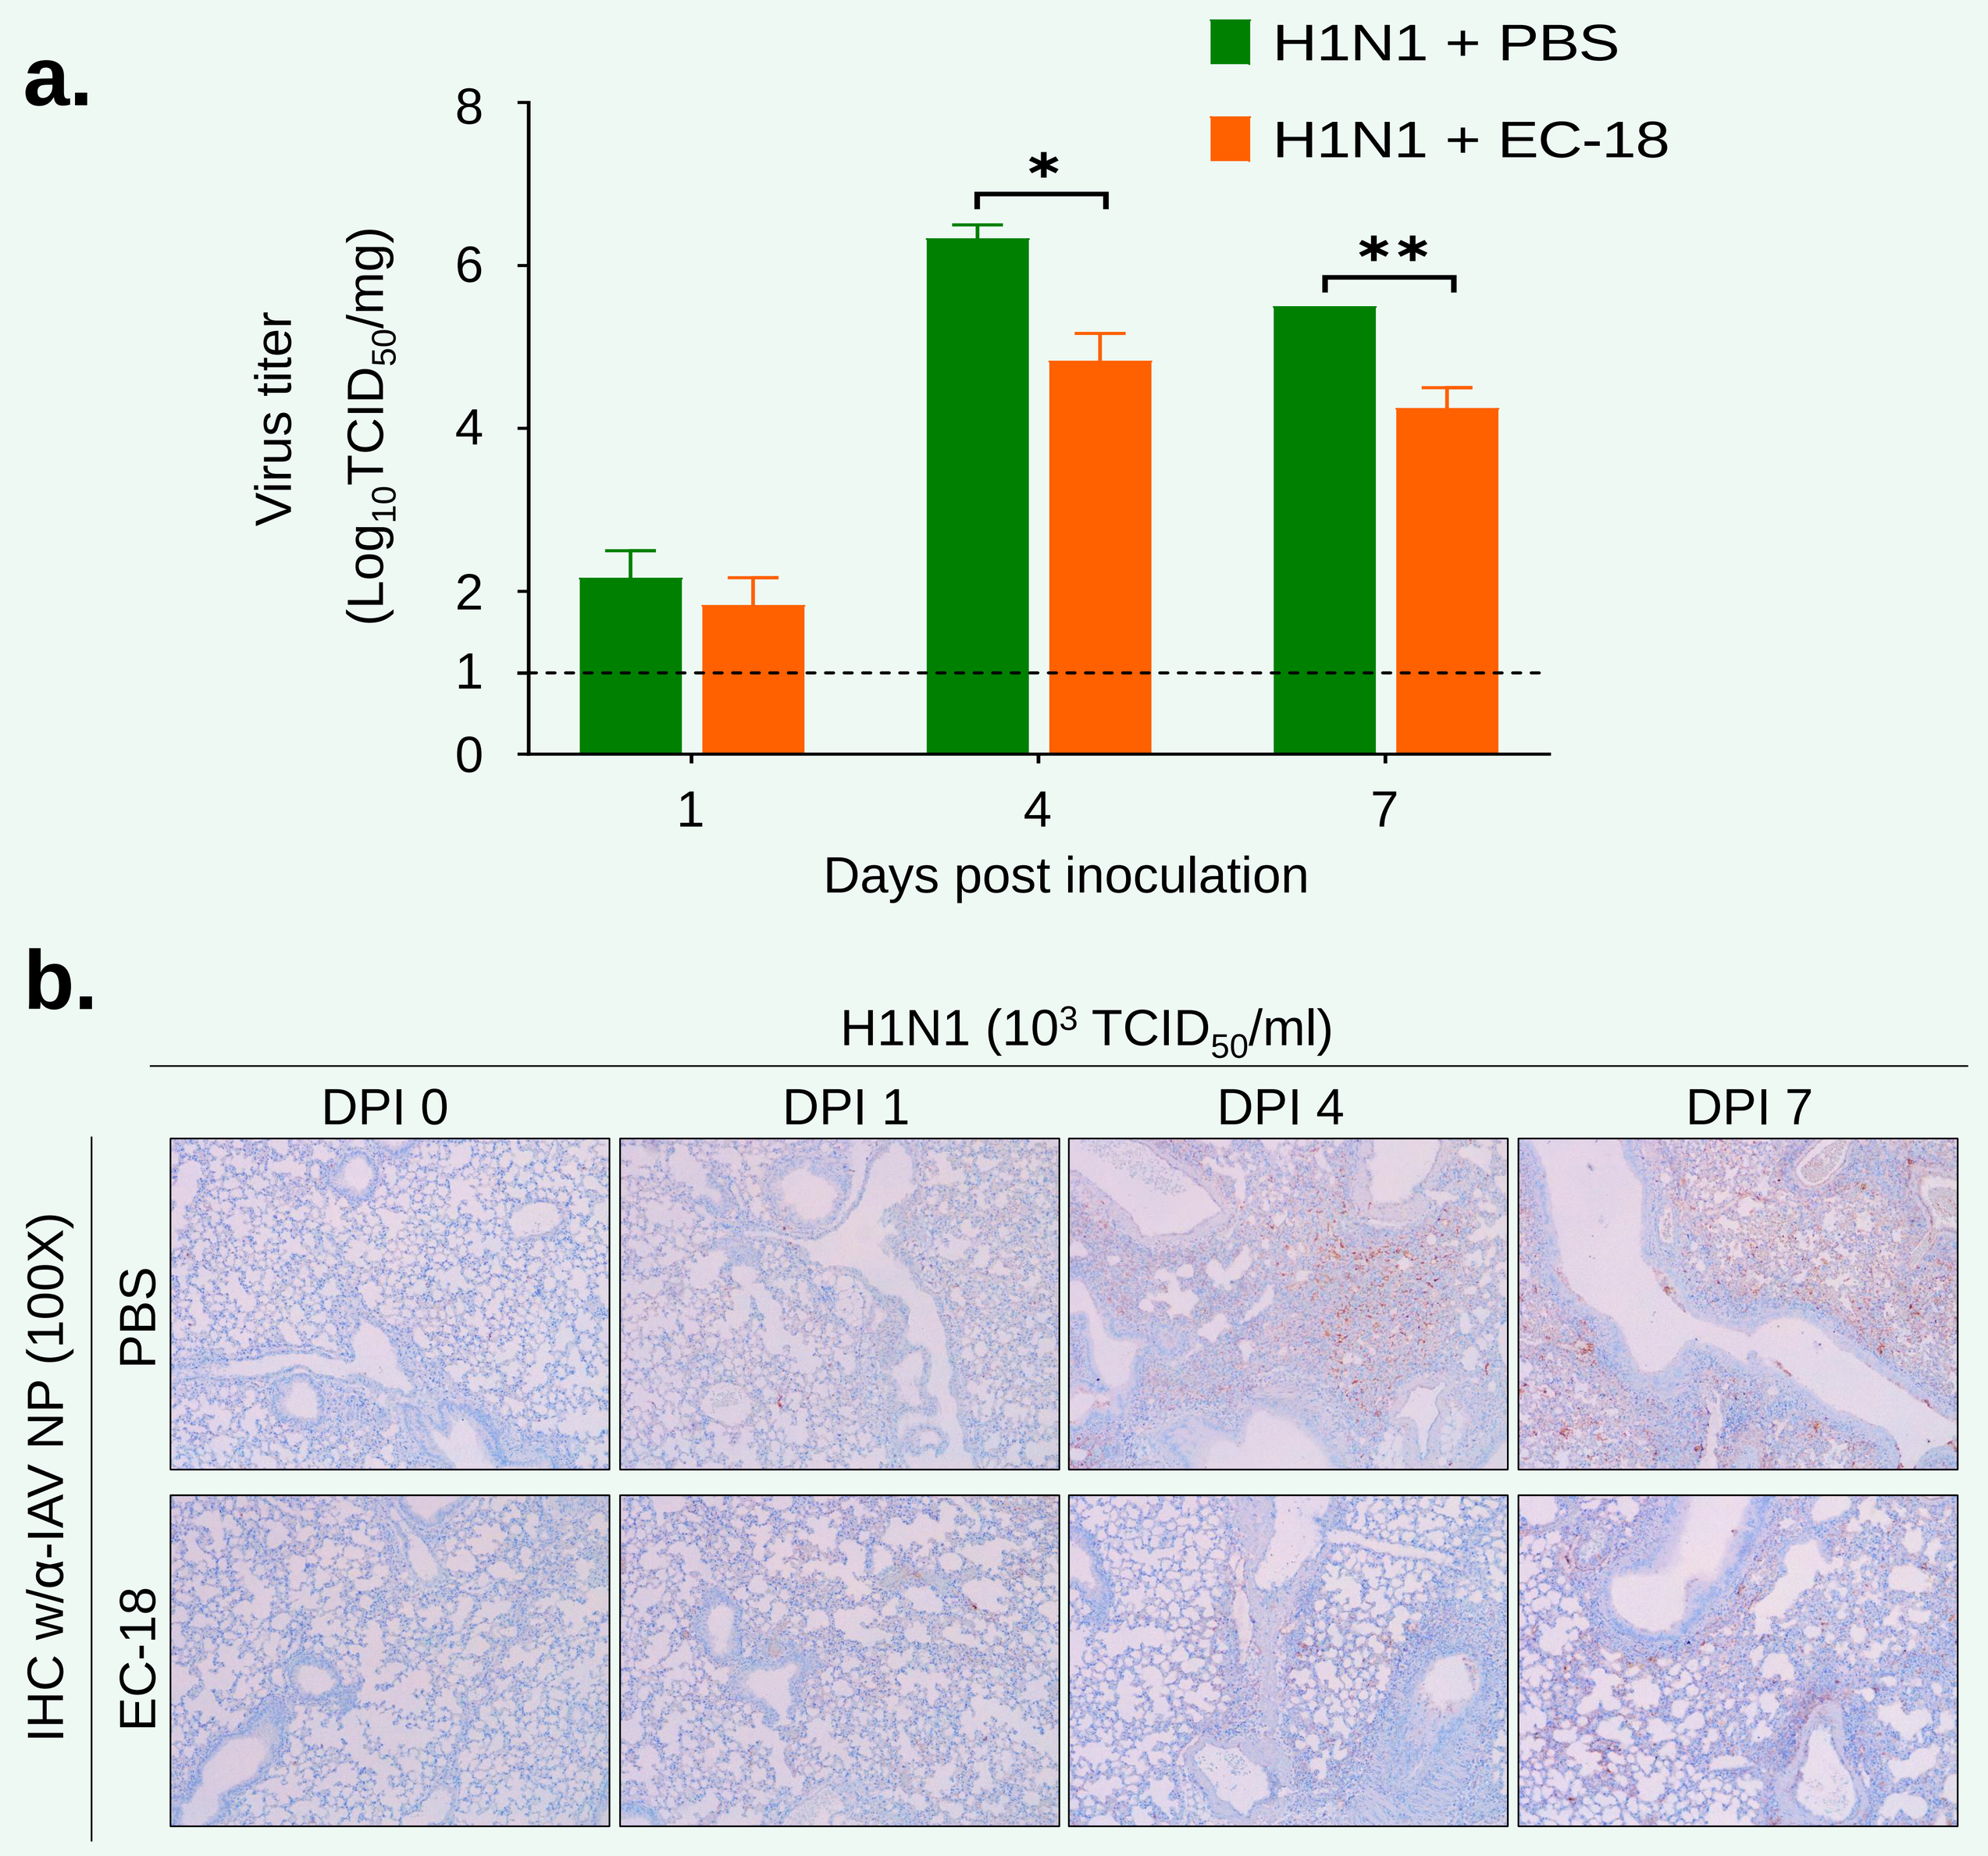
a.
H1N1 + PBS
H1N1 + EC-18
Virus titer
($Log_{10}TCID_{50}$/mg)
Days post inoculation
b.
H1N1 (10^3 $TCID_{50}$/ml)
DPI 0
DPI 1
DPI 4
DPI 7
IHC w/α-IAV NP (100X)
PBS
EC-18

Viral titers in lung of mice infected with H1N1 influenza A virus. (a) Virus titers in lung homogenate were determined by performing hemagglutination (HA) assay. **(b)** IAV antigen in lung sections was detected by performing IHC staining using anti-influenza A nucleoprotein (NP) mAb. $*p < 0.05$, $**p < 0.01$ vs. H1N1 + PBS.

3. EC-18 attenuates inflammatory cell infiltration and chemokine expression in BALF of IAV- infected mice.

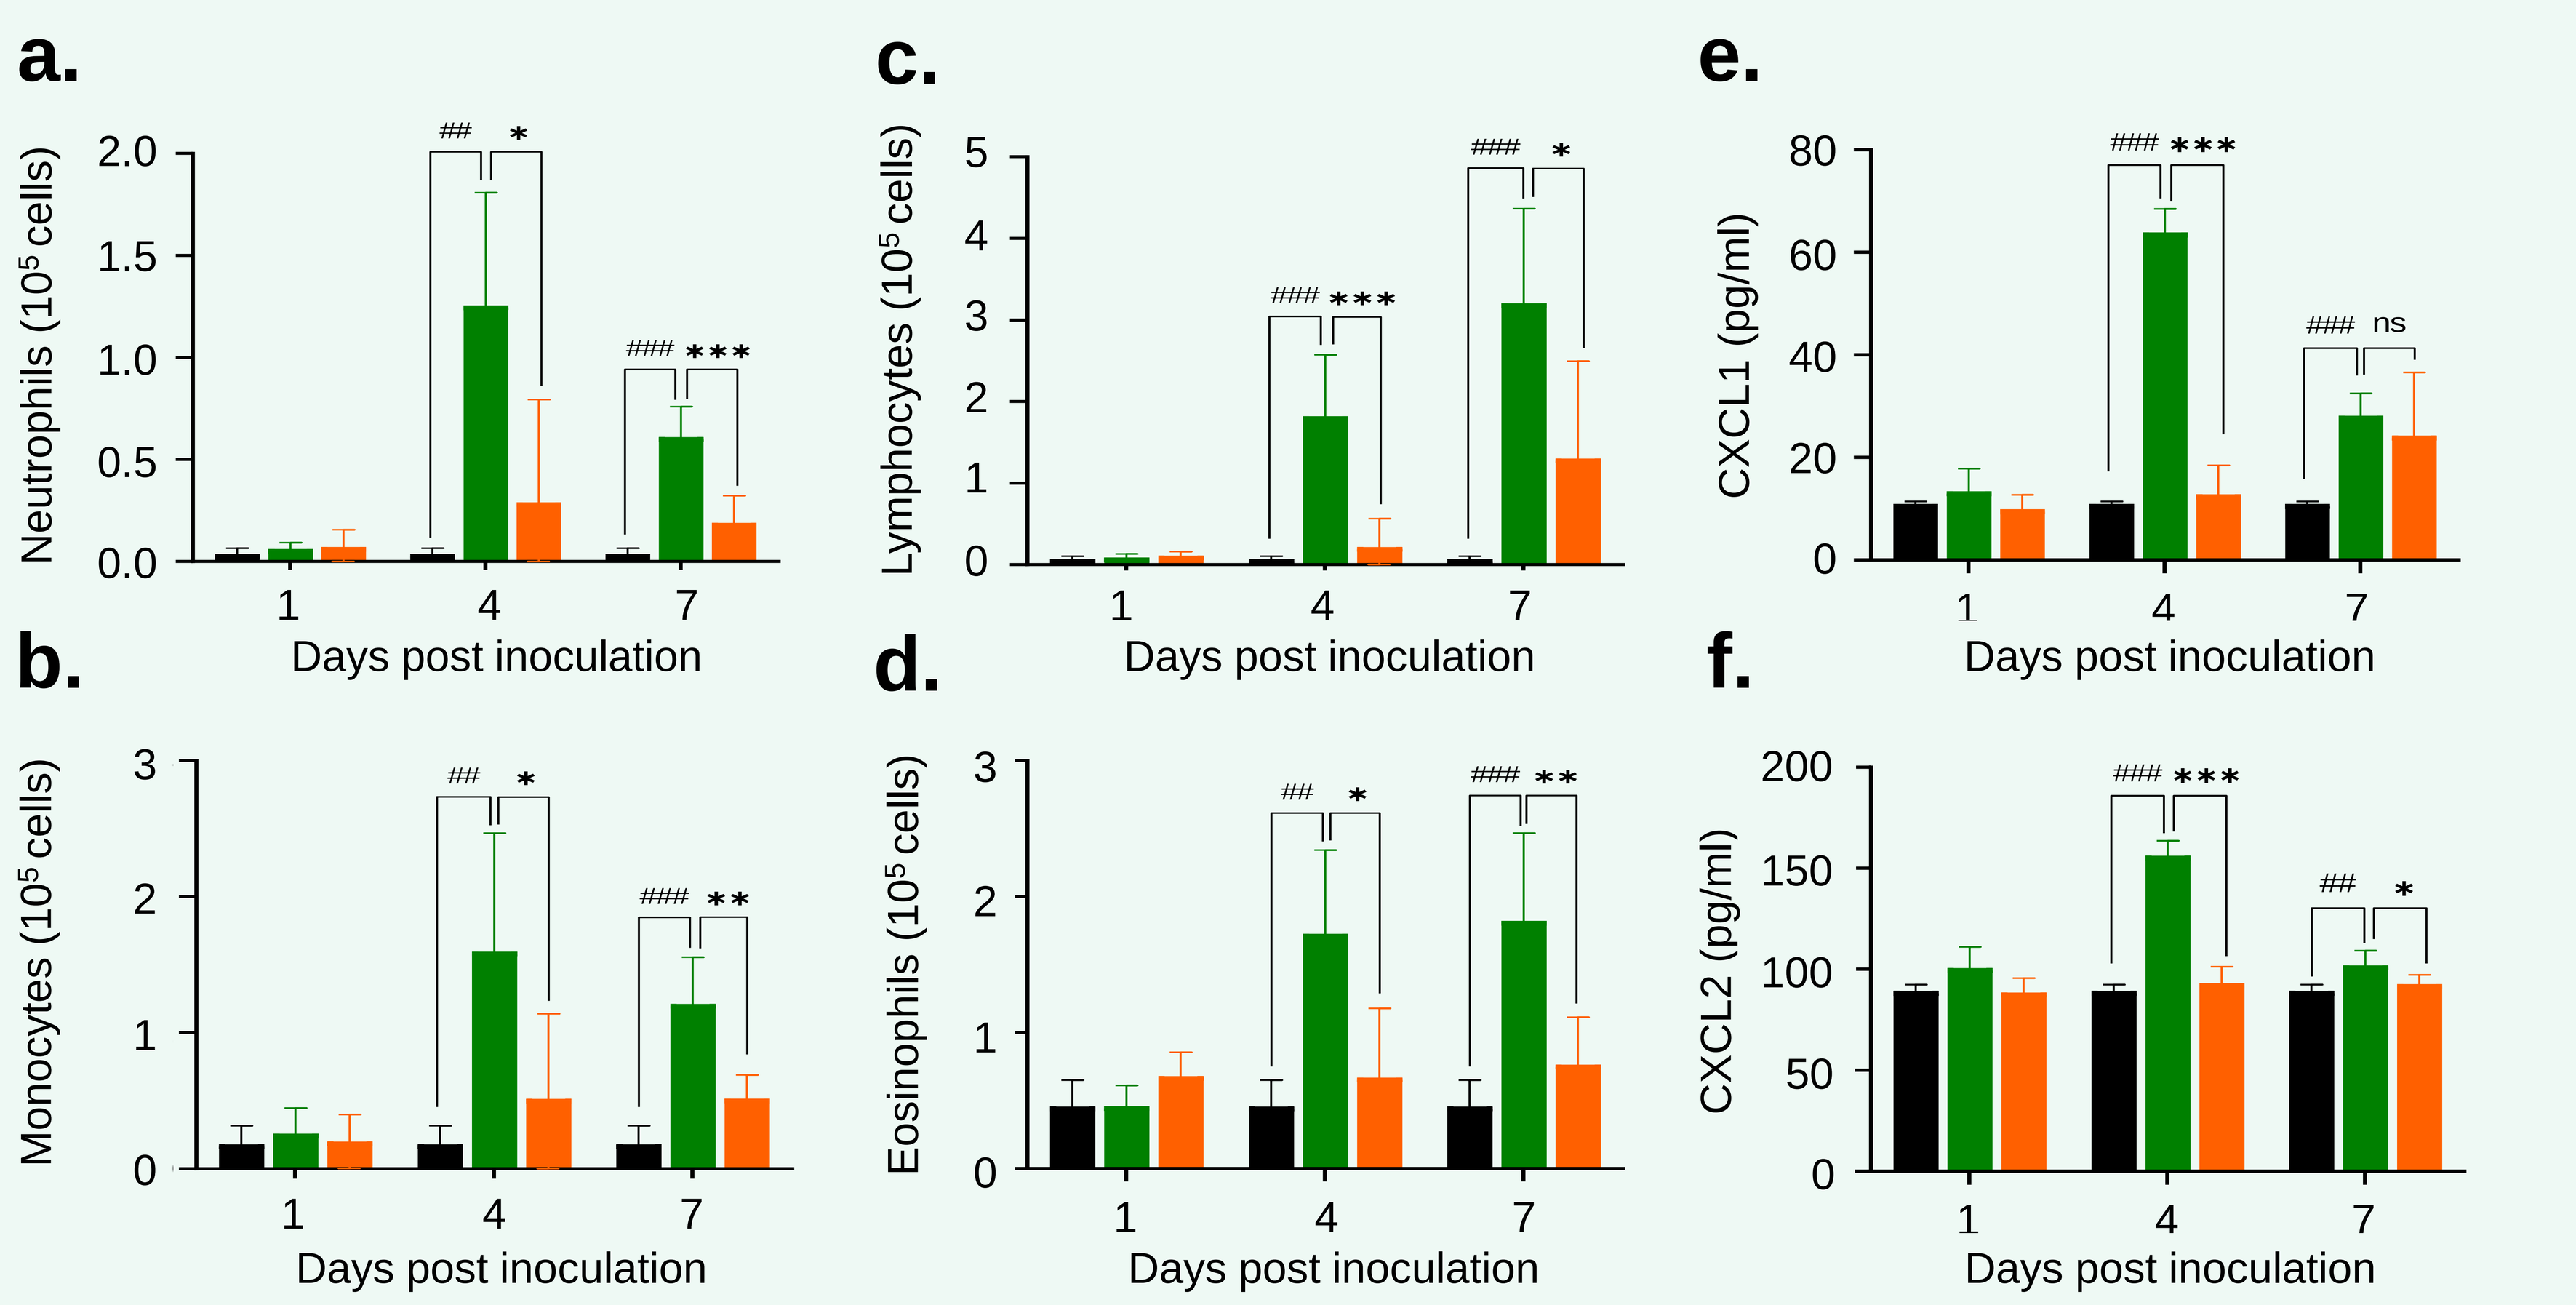
Uninfected
H1N1 + PBS
H1N1 + EC-18
a.
Neutrophils (10^5 cells)
b.
Monocytes (10^5 cells)
c.
Lymphocytes (10^5 cells)
d.
Eosinophils (10^5 cells)
e.
CXCL1 (pg/ml)
f.
CXCL2 (pg/ml)
Days post inoculation

Inflammatory profile in lung of mice following IAV infection. The mice were infected with a sub-lethal dose of H1N1 IAV, and BALF was harvested at days 1, 4, 7 post inoculation. Absolute numbers of **(a)** neutrophils **(b)** monocytes **(c)** lymphocytes **(d)** eosinophils in BALF. The protein levels of **(e)** CXCL1 and **(f)** CXCL2 in BALF. $*p < 0.05$, $**p < 0.01$ vs. H1N1 + PBS.

4. EC-18 attenuates inflammatory lung injury in IAV-infected mice.

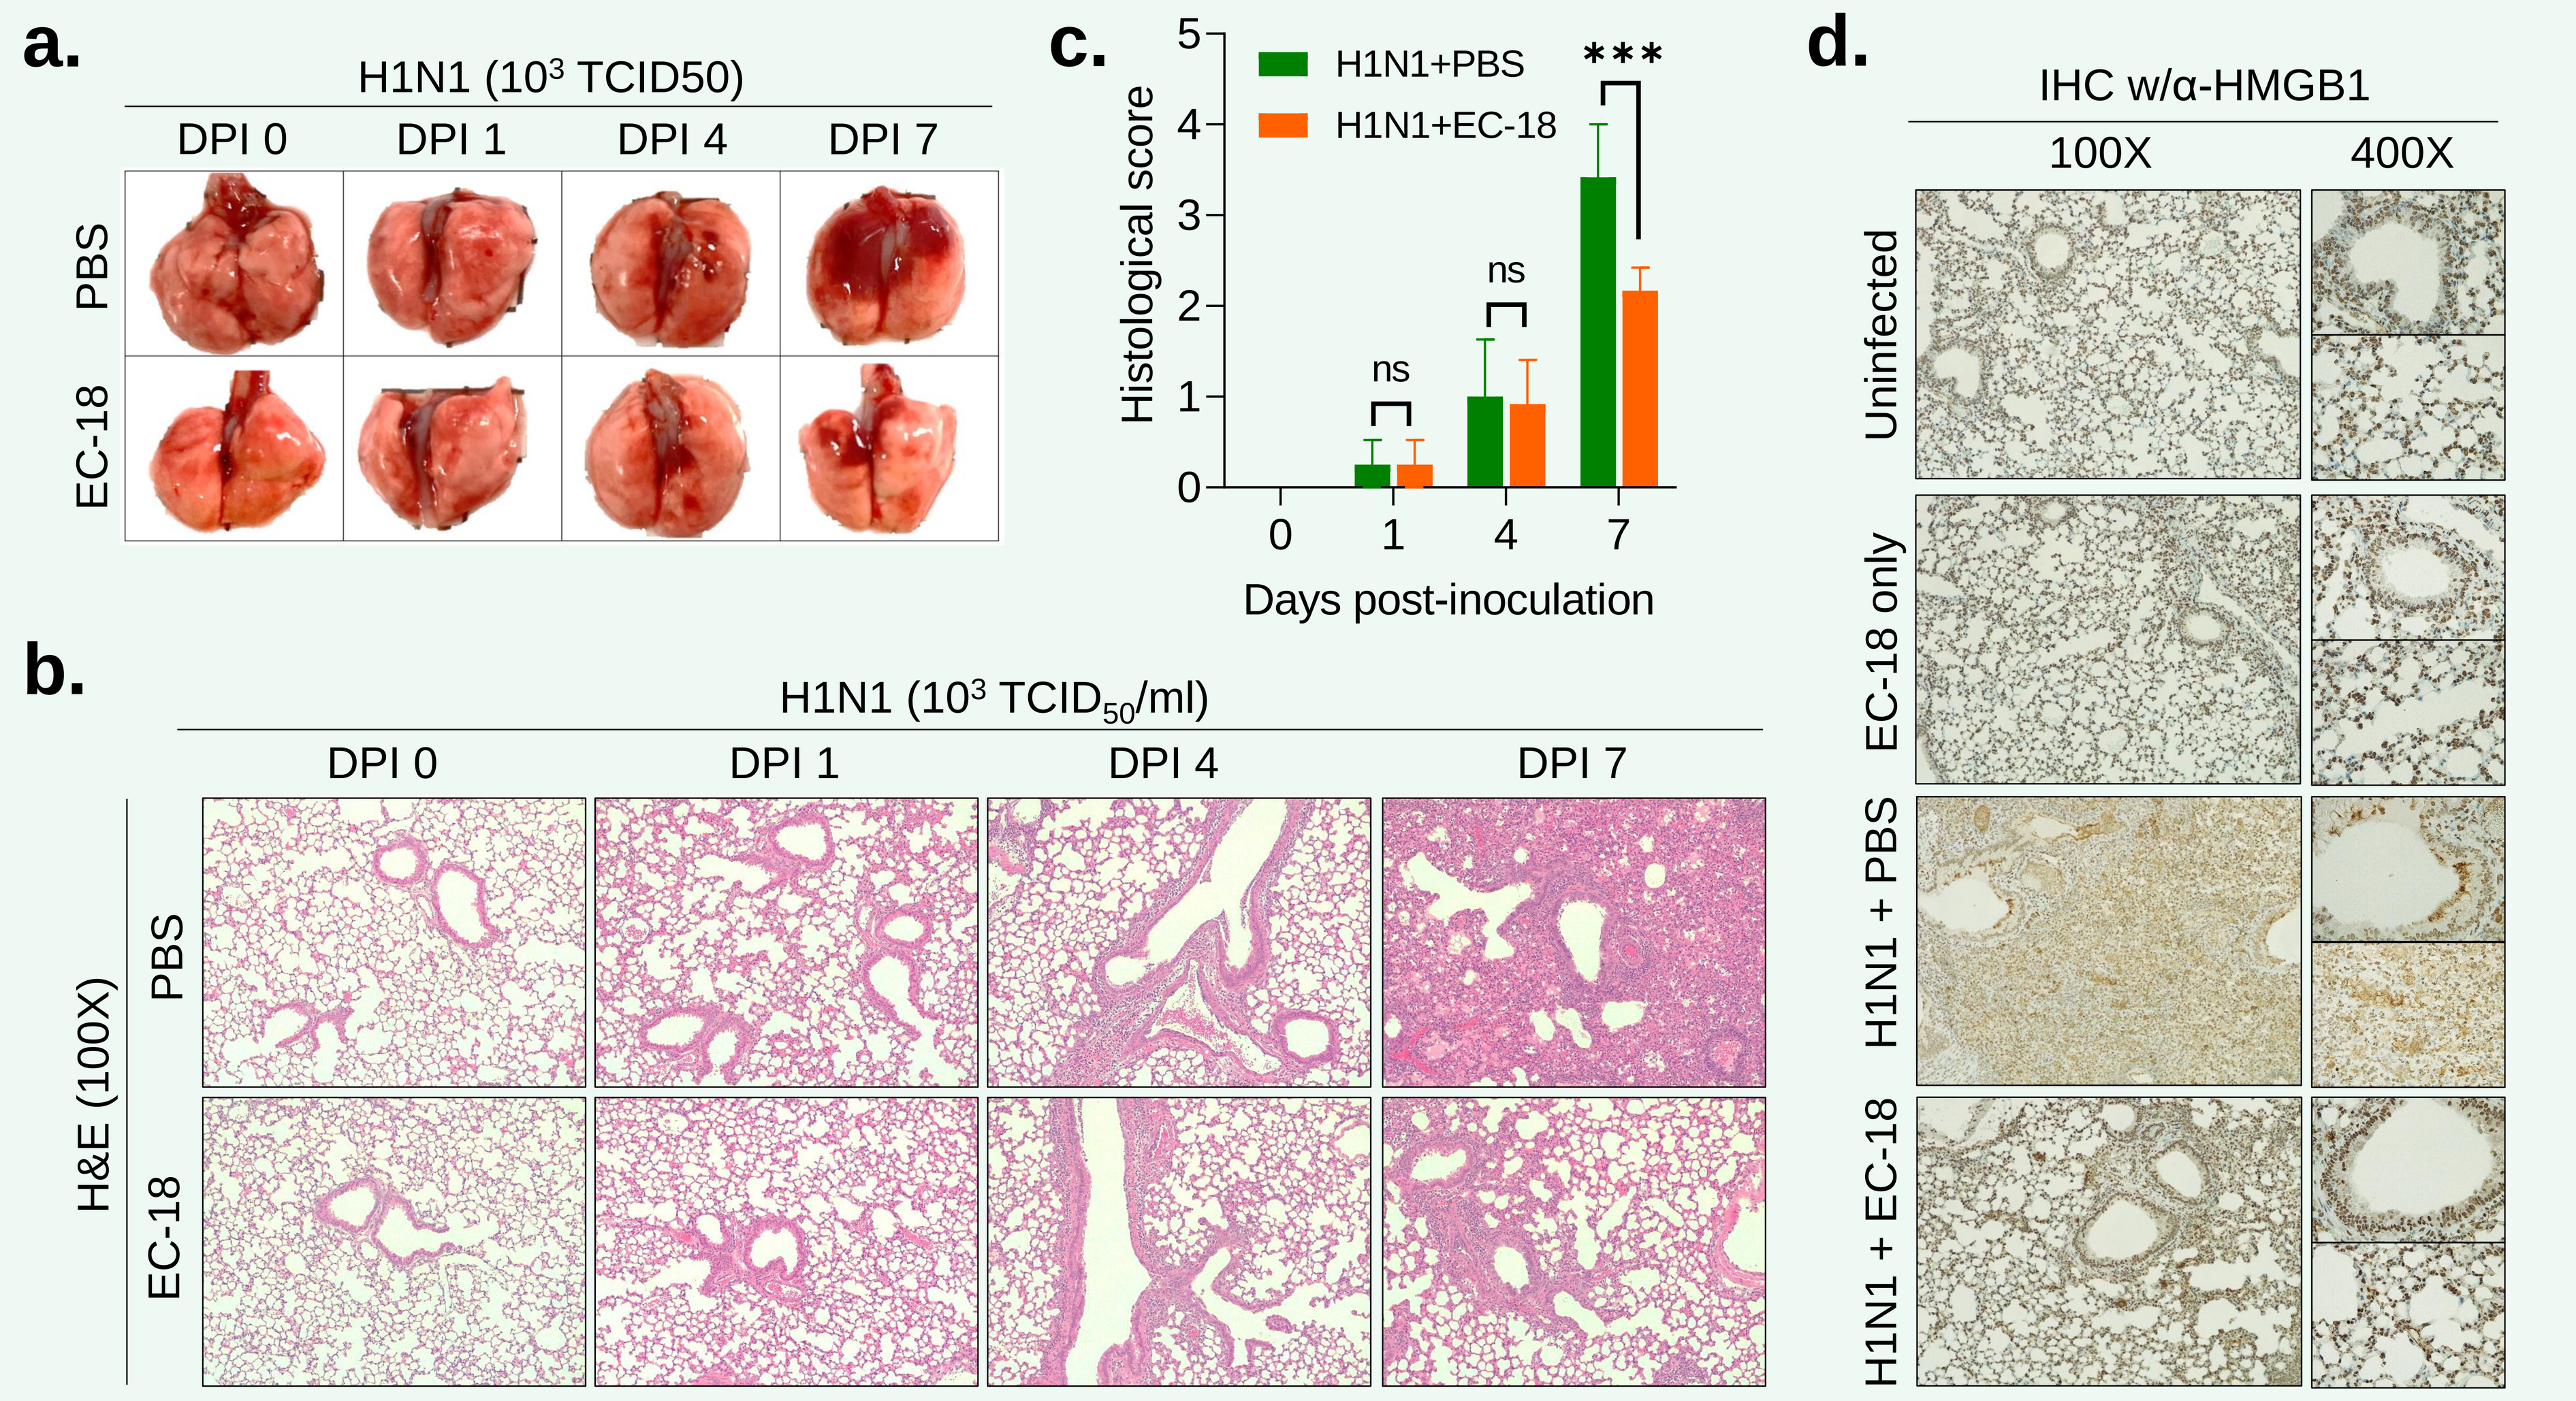
a.
H1N1 (10^3 TCID50)
DPI 0
DPI 1
DPI 4
DPI 7
PBS
EC-18
c.
H1N1+PBS
H1N1+EC-18
Histological score
Days post-inoculation
d.
IHC w/α-HMGB1
100X
400X
Uninfected
EC-18 only
H1N1 + PBS
H1N1 + EC-18
b.
H1N1 (10^3 $TCID_{50}$/ml)
H&E (100X)

Lung damage of mice following IAV infection. (a) Photographs of lungs **(b)** Lung injury was assessed by hematoxylin and eosin staining. **(c)** Histological scoring of lung injury. **(d)** Release of HMGB1 to extracellular space was determined by IHC staining. $***p < 0.001$ vs. H1N1 + PBS.

RESULT 2 : SARS-CoV-2 virus

1. Anti-viral effect of EC-18 on SARS-CoV-2 virus infection *In-vitro*.

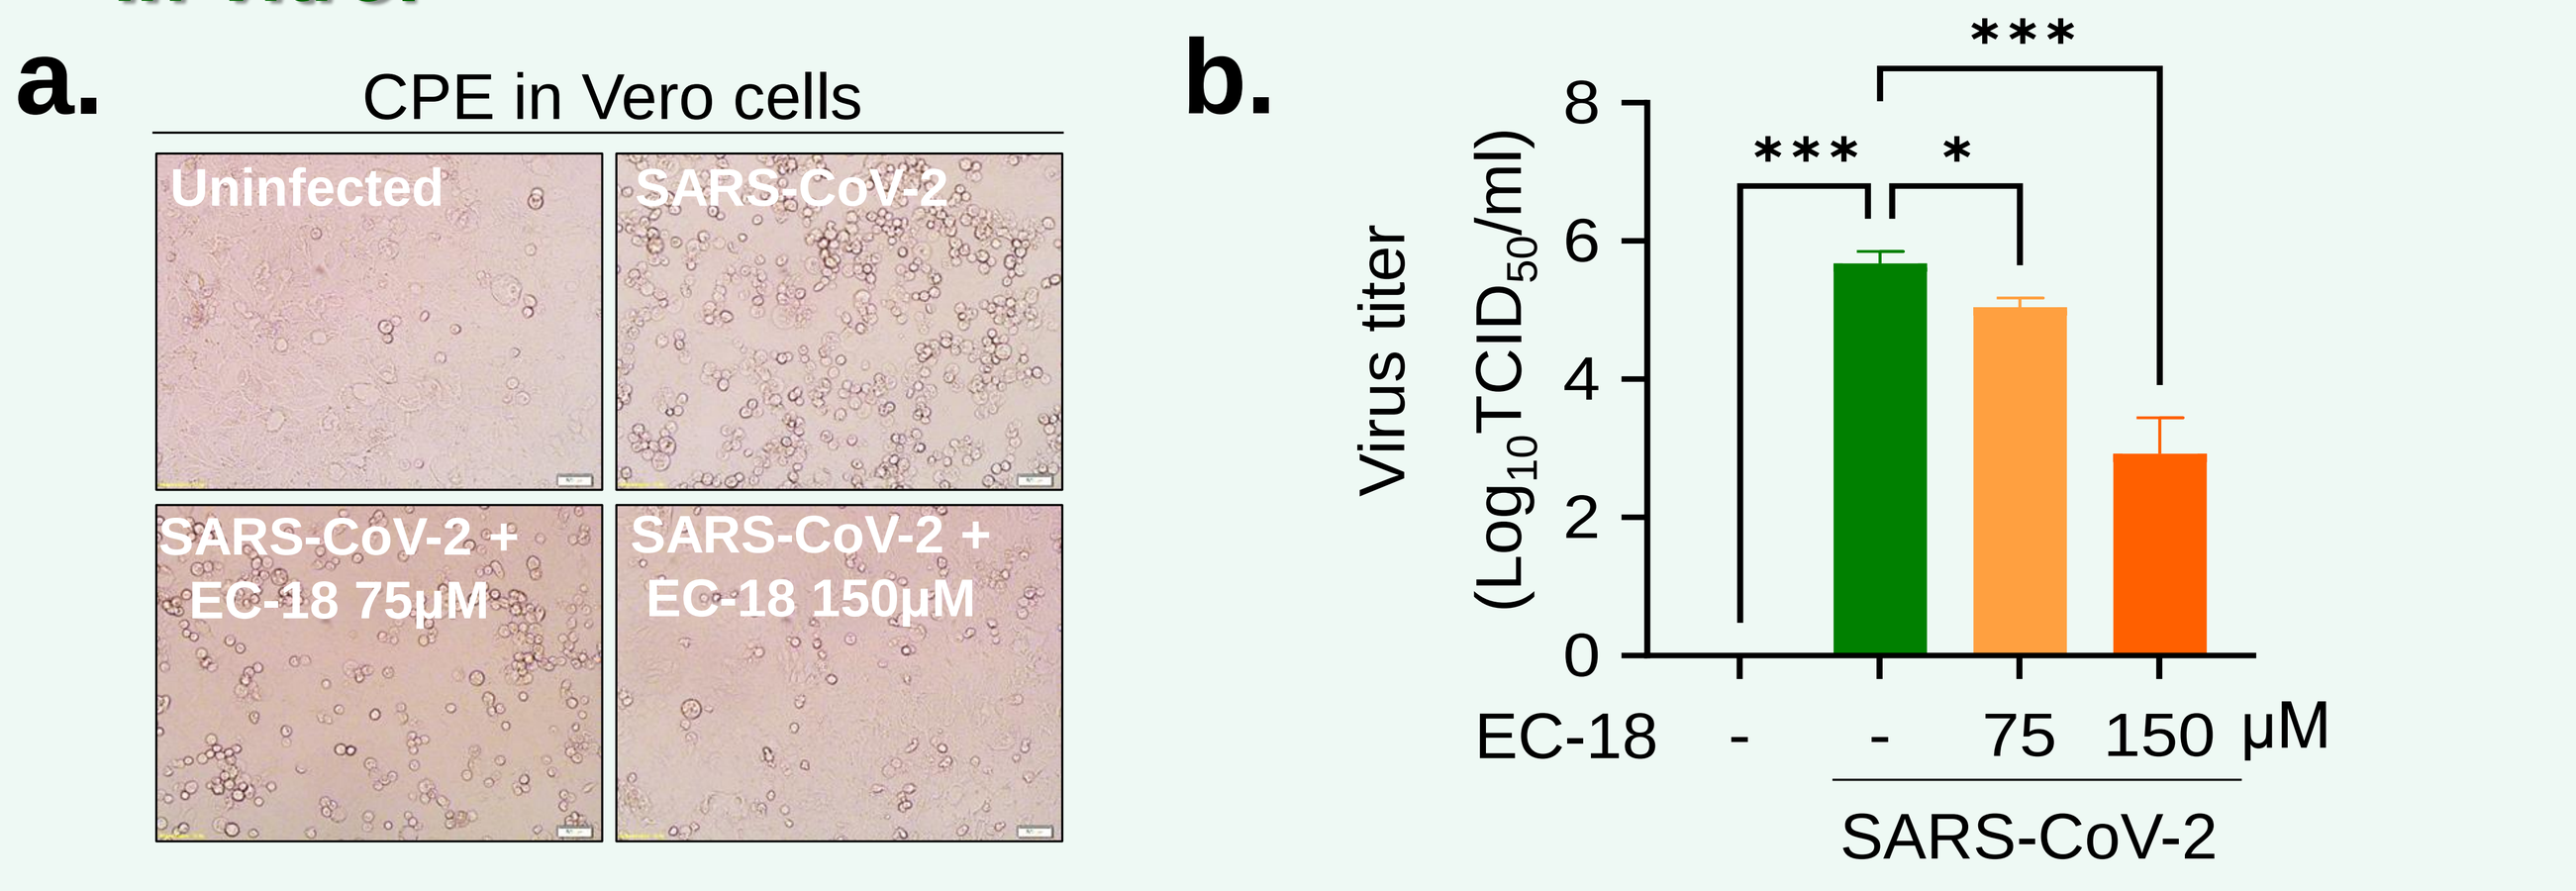
a.
CPE in Vero cells
Uninfected
SARS-CoV-2
SARS-CoV-2 + EC-18 75μM
SARS-CoV-2 + EC-18 150μM
b.
Virus titer
($Log_{10}TCID_{50}$/ml)
EC-18
75
150 μM
SARS-CoV-2

Cytopathic effect (CPE) and Viral titers in Vero cells infected with SARS-CoV-2 virus. (a) Images of CPE from SARS-CoV-2 infection in Vero cells. Cells were infected with 10^2 $TCID_{50}$/ml of the virus for 1h, then the medium was replaced with virus-free and EC-18 containing medium. After 72h, the images were obtained. **(b)** virus titer in the cell supernatants was determined by HA assay. $*p < 0.05$, $***p < 0.001$ vs. SARS-CoV-2 only.

2. EC-18 decreases chemokine expression in lung of mice infected with SARS-CoV-2.

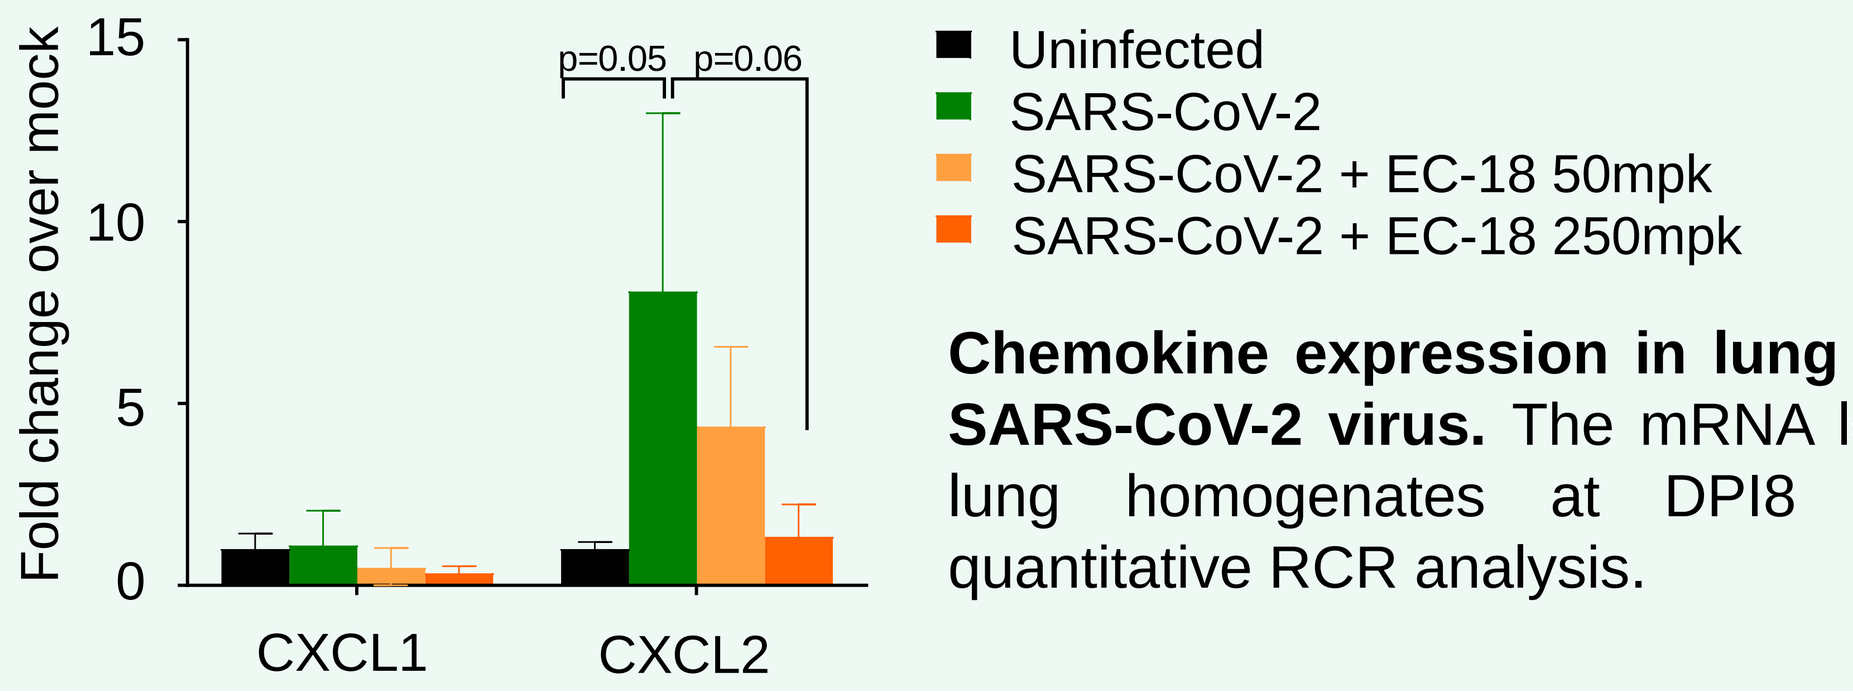
Fold change over mock
p=0.05
p=0.06
CXCL1
CXCL2
Uninfected
SARS-CoV-2
SARS-CoV-2 + EC-18 50mpk
SARS-CoV-2 + EC-18 250mpk

Chemokine expression in lung of mice infected with SARS-CoV-2 virus. The mRNA levels of chemokines in lung homogenates at DPI8 were determined by quantitative RCR analysis.

3. EC-18 attenuates inflammatory lung injury and virus titer in lung of mice infected with SARS-CoV-2.

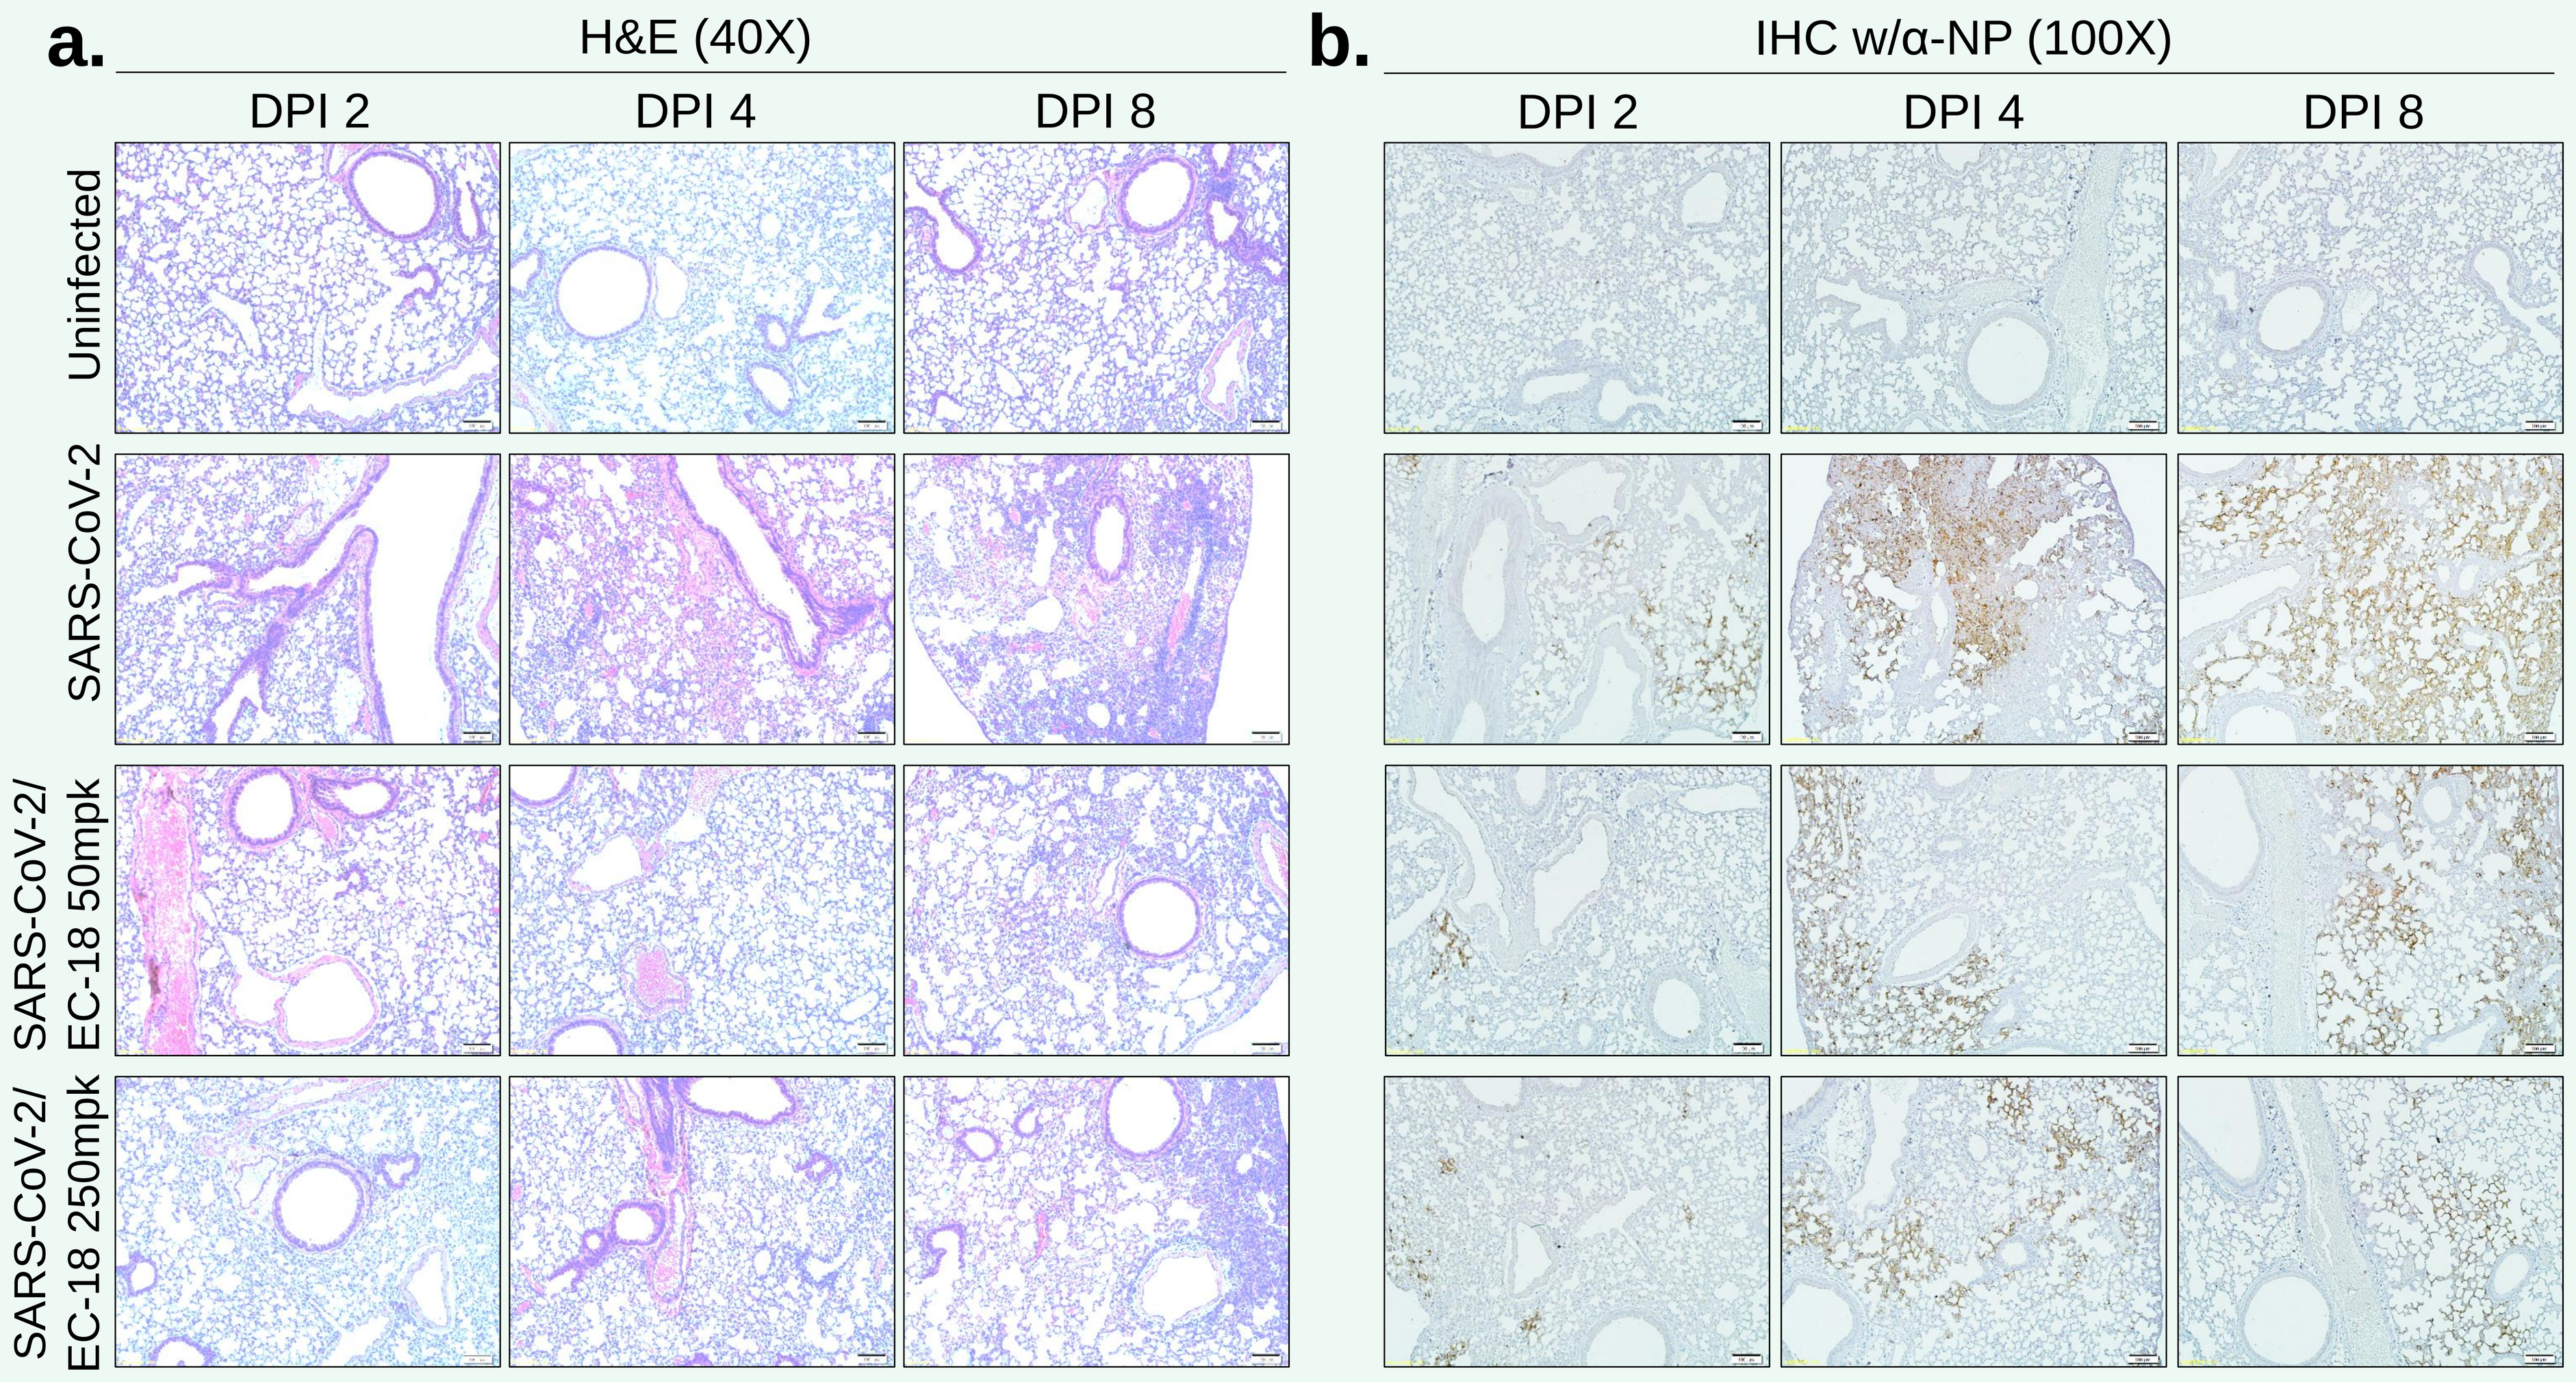
a.
H&E (40X)
b.
IHC w/α-NP (100X)
DPI 2
DPI 4
DPI 8
Uninfected
SARS-CoV-2
SARS-CoV-2/ EC-18 50mpk
SARS-CoV-2/ EC-18 250mpk

Inflammatory lung damage and invaded virus in lung following infection. (a) Lung injury was assessed by hematoxylin and eosin staining. **(b)** SARS-CoV-2 antigen in lung sections was detected by performing IHC staining using nucleoprotein (NP) mAb.